The University of Manchester

Université Claude Bernard Lyon

Modelling of insulin secretion : from beta-cell physiology to sulphonylureas-mediated insulin secretion

D. Cronier[1,2], E. Grenier[3], M. Lecomte[4], V. Autier[4], B. Ladstetter[4], L.J. Aarons[2], and J.P. Boissel[1]

1: RTH School of Medicine, University Claude Bernard Lyon, France ; 2: School of Pharmacy and Pharmaceutical Sciences, University of Manchester, U.K 3: Ecole Normale Supérieure, Lyon, France ; 4: MERCK Santé Research Center, Chilly mazarin, France
5: Global Non Clinical DMPK, MERCK KGAa, Grafing, Germany

Study aim

1. To develop an integrated model describing the pancreatic physiological response to glucose, including beta-cell physiology and insulin secretion by an isolated pancreas
2. To use this modelling framework to investigate the effect of sulphonylureas (SU) on beta-cell physiology and insulin secretion

Model Development

The endocrine pancreas is assumed to be constituted by an homogeneous population of beta-cells behaving strictly identically. The global model describing pancreatic insulin secretion is therefore made up of three sub models describing the major cellular events underlying the insulin response to glucose, i.e. the metabolic, electrophysiological and exocytotic events.

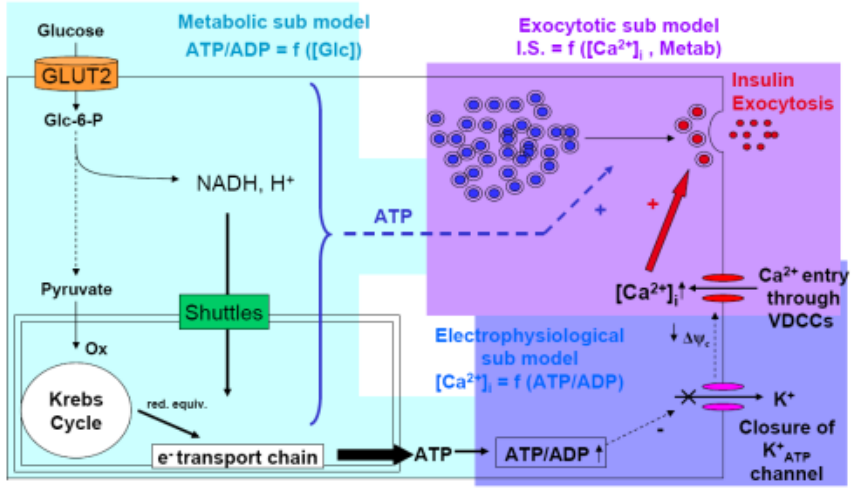

Glucose
GLUT2
Glc-6-P
Metabolic sub model
ATP/ADP = f ([Glc])
Exocytotic sub model
I.S. = f ([Ca2+]i , Metab)
Insulin Exocytosis
NADH, H+
ATP
Pyruvate
Shuttles
Ox
Krebs Cycle
red. equiv.
e- transport chain
ATP
ATP/ADP
[Ca2+]i
Ca2+ entry through VDCCs
Electrophysiological sub model
[Ca2+]i = f (ATP/ADP)
K+
Closure of K+ATP channel

Fig. 1: global integrated model.

Metabolic sub model

The metabolic sub model is an **Inside Fluid Compartment Model**. Each compartment is considered an homogenous region in which compounds diffuse freely and instantly. Only slow reactions are described by means of differential equations describing the conservation of mass, where enzyme rates are described by means of the Hill equation.

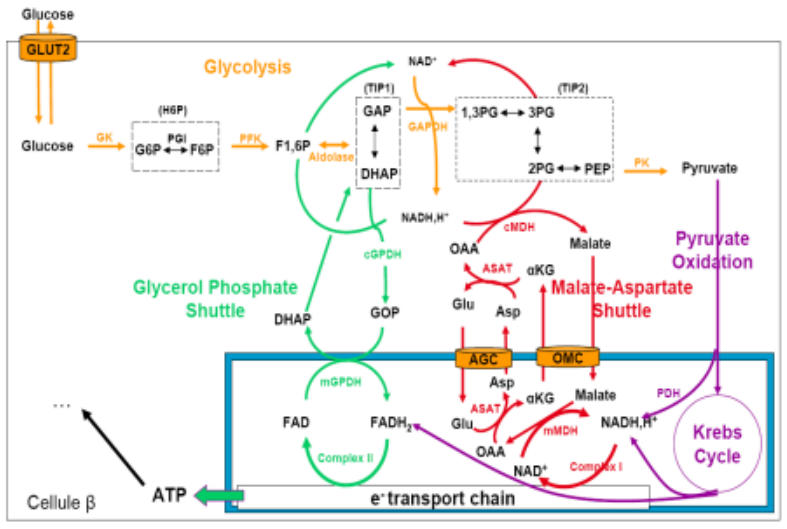

Glucose
GLUT2
Glycolysis
Glycerol Phosphate Shuttle
Malate-Aspartate Shuttle
Pyruvate Oxidation
Krebs Cycle
Cellule β
ATP
e- transport chain

Fig. 2: metabolic sub model.

$$A \xrightarrow{E1} B \xrightarrow{E2} C$$

$$\frac{dB}{dt} = \varphi_A^{E1} - \varphi_B^{E2}$$

$$\varphi_x^E = \frac{V_{m,x}^E \cdot [X]^{\gamma}}{(K_{0.5}^E)^{\gamma} + [X]^{\gamma}}$$

Electrophysiological sub model

The electrophysiological model is a **Parallel Conductance Model** where the membrane is treated as an electronic circuit. The dielectric properties are represented by a capacitance and the parallel conductances correspond to the different ion channels and electrogenic carriers.

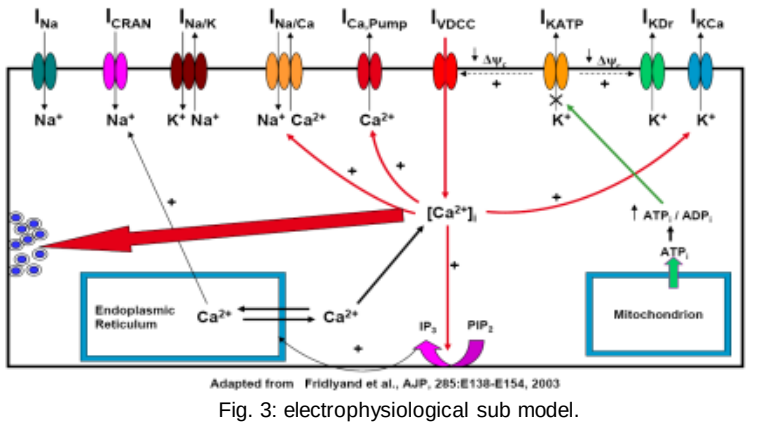

INa
ICRAN
INa/K
INa/Ca
ICa,Pump
IVDCC
IKATP
IKDr
IKCa
[Ca2+]i
Endoplasmic Reticulum
Mitochondrion
Adapted from Fridlyand et al., AJP, 285:E138-E154, 2003

Fig. 3: electrophysiological sub model.

$$\frac{dV}{dt} = -\frac{1}{C} \cdot \sum I_{j,i}$$

with $I_{j,i} = g_{m,j} \cdot (V - V_i)$

and $V_i = \frac{RT}{zF} \ln\left(\frac{[\,]_e}{[\,]_i}\right)$

Exocytotic sub model

The exocytotic model is a **Compartmental Model**. The global insulin content is split into two separate pools. The mature insulin is secreted from an immediately releasable pool (IRP) by means of a calcium activation of granule fusion. The refilling of the IRP is dependent on the delayed ATP-dependent activation of the transfer and maturation of granules from a Reserve Pool (RP) containing immature granules.

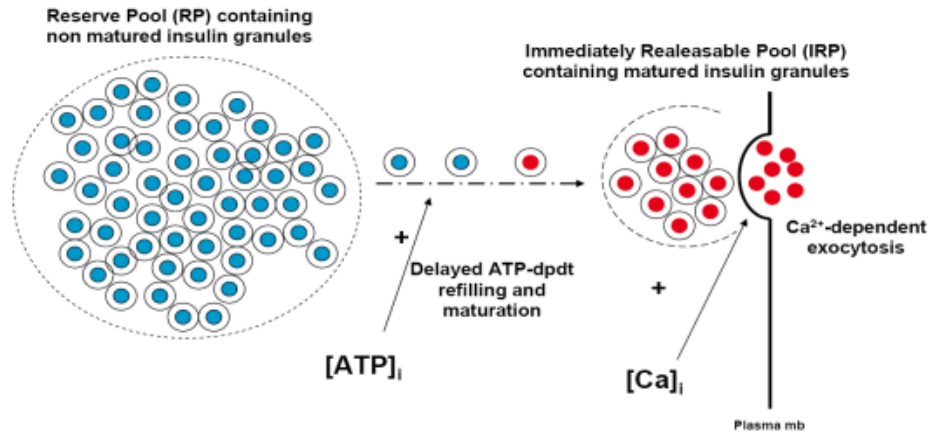

Reserve Pool (RP) containing non matured insulin granules
Immediately Realeasable Pool (IRP) containing matured insulin granules
Delayed ATP-dpdt refilling and maturation
Ca2+-dependent exocytosis
[ATP]i
[Ca]i
Plasma mb

Fig. 4: exocytotic sub model.

Results

Physiological insulin response to glucose

A repeated glucose stimulation on an isolated rat pancreas by 7, 10, and 16 mM glucose was simulated.

Beta-cell physiology
The model could reproduce the evolution of the key variables describing the beta-cell physiological response to glucose. The ATP to ADP ratio is increased in a glucose-dependent manner and follows an oscillating pattern until 16 mM glucose. The plasma membrane potential is characterized by a bursting pattern increasing in frequency until continuous spiking at 16 mM glucose . The cytosolic calcium is also characterized by an oscillating pattern following plasma membrane potential.

Insulin secretion
The model was able to reproduce the biphasic pattern of insulin secretion by an isolated pancreas. The glucose-dependent activation of calcium oscillations led to the release of the insulin contained in the IRP, responsible for the first phase the extent of which is commensurate to the level of glucose stimulus. The elevation of the ATP concentration gives rise to the delayed rising second phase.

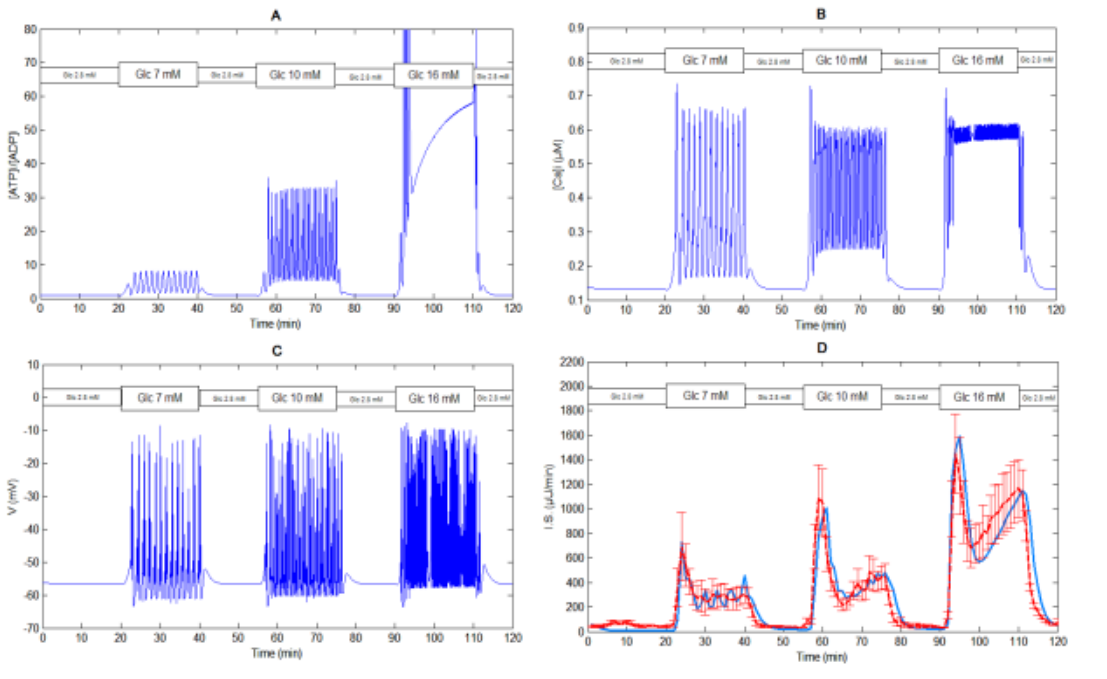

A
B
C
D
Glc 7 mM
Glc 10 mM
Glc 16 mM
Time (min)

Fig. 5: Simulation of a repeated glucose stimulation of an isolated perfused rat pancreas. A: simulated time course of the cytosolic ATP to ADP ratio. B : simulated time course of cytosolic calcium concentration. C: simulated time course of plasma membrane potential. D: experimental (red line) and simulated (blue line) time courses of insulin secretion.

Insulin response to sulphonylureas

A 90% inhibition of the K_{ATP} channel, achieved a 10 µM concentration of Gliclazide at 2.8 mM Glc on an isolated rat pancreas, was simulated. The inihibition of the K_{ATP} channel was simulated by means of a 90% decrease in KATP channel conductance.

Beta-cell physiology
The model can reproduce the uncoupling between the metabolic and electrophysiological events characterizing the effect of SUs upon beta-cells. No increase in the ATP to ADP ratio can be observed whereas both the membrane potential and cytosolic calcium are increased and follow an oscillating pattern characterizing insulin secretion.

Insulin secretion
The activation of the distal part of the cascade triggers the calcium oscillations responsible for the first phase of pancreatic insulin secretion profile. However, the lack of activation of glucose metabolism as well as the distal activation of the insulin secretion cascade led to the absence of second phase and therefore to a monophasic pattern.

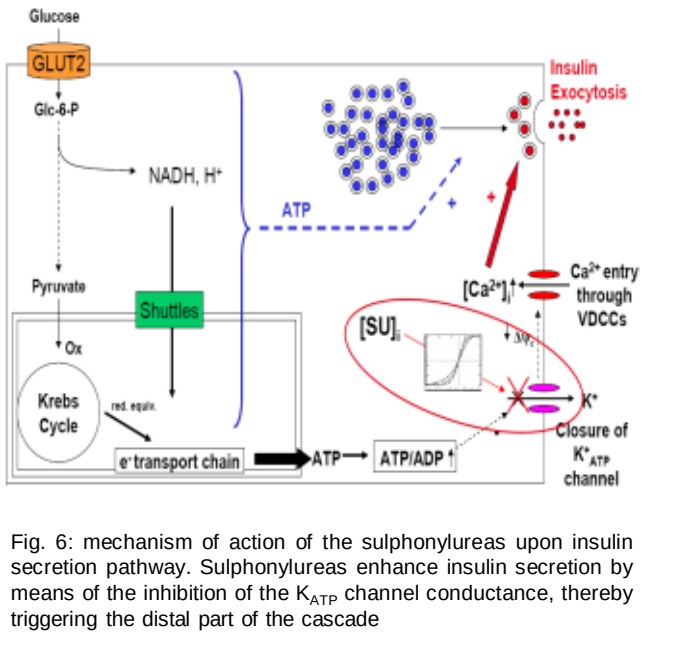

Glucose
GLUT2
Glc-6-P
NADH, H+
ATP
Pyruvate
Shuttles
Ox
Krebs Cycle
red. equiv.
e- transport chain
ATP
ATP/ADP
Insulin Exocytosis
[Ca2+]i
Ca2+ entry through VDCCs
[SU]i
K+
Closure of K+ATP channel

Fig. 6: mechanism of action of the sulphonylureas upon insulin secretion pathway. Sulphonylureas enhance insulin secretion by means of the inhibition of the K_{ATP} channel conductance, thereby triggering the distal part of the cascade

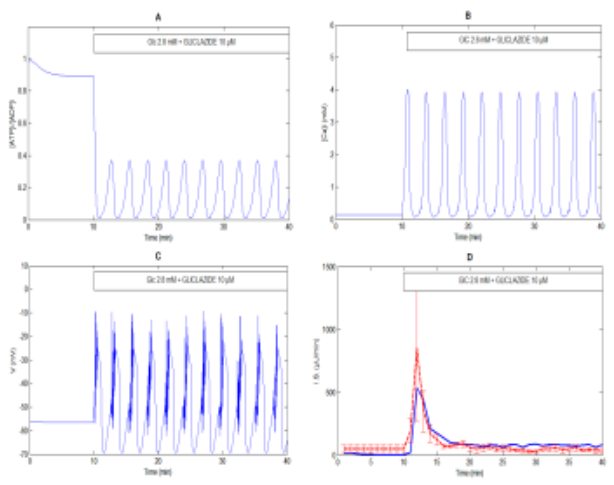

A
B
C
D

Fig. 7: Simulation of of the stimulation of perfused isolated rat pancreas with 10 µM Gliclazide and 2.8 mM Glucose. A: simulated time course of the ATP to ADP ratio. B: simulated time course of the cytosolic calcium concentration. C: simulated time course of plasma membrane potential. D: corresponding experimental (red line) and simulated (blue line) time course of insulin secretion

Conclusion

The global pancreatic integrated model could reproduce both the physiological and SU-mediated insulin responses.
Under glucose stimulation the model could reproduce the evolution of the main metabolic and electrophysiological variables as well as the biphasic insulin secretion pattern .
Under sulphonylurea stimulation, the model could reproduce the uncoupling between metabolism and the distal part of the insulin secretion cascade, therefore leading to a monophasic insulin secretion pattern .